Zorevunersen (STK-001) demonstrates potential for disease modification, including reductions in seizures and improvements in cognition and behavior in children and adolescents with Dravet syndrome

Poster 2.379

Linda Laux[1], J. Helen Cross[2], Joseph Sullivan[3], Archana Desurkar[4], Colin M. Roberts[5], John M. Schreiber[6], Kelly G. Knupp[7], M. Scott Perry[8], James W. Wheless[9], Elaine C. Wirrell[10], Charlene Brathwaite[11], Carrie Condon[11], Ann Dandurand[11], Jessie Lynch[11], James Stutely[11], Fei Wang[11], Meena[11], Kimberly A. Parkerson[11], Barry Ticho[11]

[1]Ann & Robert H. Lurie Children's Hospital of Chicago, Chicago, IL, USA; [2]Great Ormond Street Hospital for Children NHS Foundation Trust, London, UK; [3]University of California, San Francisco, CA, USA; [4]Sheffield Children's Hospital NHS Foundation Trust, Sheffield, UK; [5]Oregon Health & Science University, Portland, OR, USA; [6]Children's National Hospital, Washington D.C., USA; [7]Children's Hospital Colorado, Aurora, CO, USA; [8]Cook Children's Medical Center, Fort Worth, TX, USA; [9]Le Bonheur Children's Hospital, University of Tennessee Health Science Center, Memphis, TN, USA; [10]Mayo Clinic, Rochester, MN, USA; [11]Stoke Therapeutics, Inc., Bedford, MA, USA.

INTRODUCTION

- Zorevunersen is an investigational antisense oligonucleotide designed to treat the underlying cause of Dravet syndrome (DS) — $Na_V1.1$ protein haploinsufficiency caused by variants of the sodium channel type 1 alpha subunit (*SCN1A*) gene[1,2]
- DS is a severe developmental and epileptic encephalopathy characterized by severe, recurrent seizures as well as significant cognitive and behavioral impairments[3–5]
- Despite treatment, 90% of children and adolescents with DS experience uncontrolled seizures, and impacts on cognition and behavior are not fully addressed by current therapies[5]
- Even with standard of care treatment with the best antiseizure medications (ASMs), patients with DS enrolled in the BUTTERFLY natural history study demonstrated ongoing high rates of seizures and fell behind in aspects of cognition and behavior over 24 months relative to their neurotypical peers[6]
- Disease-modifying therapies targeting the pathophysiology of DS are needed to improve short- and long-term outcomes
- The MONARCH and ADMIRAL Phase 1/2a studies evaluated the safety and clinical effects of zorevunersen in patients with DS already receiving standard of care ASMs

METHODS

- MONARCH and ADMIRAL were open-label, multi-center studies conducted in the USA and UK, respectively (Figure 1)
- In both studies, patients received zorevunersen at their assigned dose level as an intrathecal slow bolus over 1 to 3 minutes

Figure 1. MONARCH and ADMIRAL study design

Primary objectives include safety and tolerability of single and multiple doses of zorevunersen, change in plasma PK, and CSF exposure
Secondary and exploratory objectives include effects of zorevunersen on percentage change from baseline in convulsive seizure frequency, overall clinical status (CGI-C/CaGI-C), QoL (EQ-VAS component of EQ-5D-Y), and cognition and behavior (Vineland-3)

Eligibility criteria
- 2–18 years of age
- Established DS diagnosis
- Documented pathogenic, likely pathogenic, or variant of uncertain significance in the *SCN1A* gene

MONARCH study dosing and cohorts:
Single ascending dose at 10, 20, 30, 45, and 70 mg with drug administration on Day 1
Multiple ascending dose at 20, 30, and 45 mg with drug administration on Days 1, 29, and 57

ADMIRAL study dosing and cohorts:
Multiple ascending dose at 30, 45, and 70 mg with drug administration on Days 1, 57, and 85
Multiple ascending dose at 70 mg with drug administration on Days 1 and 57

CaGI-C, Caregiver Global Impression of Change; CGI-C, Clinical Global Impression of Change; CSF, cerebrospinal fluid; DS, Dravet syndrome; EQ-5D-Y, EuroQol-Five dimensions-Youth; EQ-VAS, EuroQol Visual Analogue Scale; PK; pharmacokinetics; QoL, quality of life; *SCN1A*, sodium channel type 1 alpha subunit; Vineland-3, Vineland Adaptive Behavior Scales, Third Edition.

Key Findings

1. Results from MONARCH and ADMIRAL Phase 1/2a studies support the **potential of zorevunersen to be the first disease-modifying medicine for DS**
2. Treatment with zorevunersen resulted in **seizure reduction and improvements in cognition and behavior, overall clinical status, and quality of life within the first 9 months** in patients already receiving best available ASMs
3. The most substantial improvements were observed in **patients treated with 70 mg zorevunersen**
4. Treatment with zorevunersen was generally **well tolerated**

RESULTS

Baseline characteristics

- Overall, 81 patients received ≥1 dose of zorevunersen across all dose levels (10–70 mg) (Table 1)
- Patients were highly refractory to available treatments, with 85% and 54% taking ≥3 and ≥4 concomitant ASMs, respectively
 - 49% of all patients were taking fenfluramine
- Median baseline convulsive seizure frequency per 28 days was 17 in clinically evaluable patients

Table 1. Summary of baseline characteristics

Parameter	MONARCH (n=62)	ADMIRAL (n=19)	ALL ENROLLED (N=81)
Age at screening, years			
Median (min, max)	10.0 (2, 18)	7.0 (3, 17)	10.0 (2,18)
Age group, n (%)			
2–12 years	35 (56.5)	11 (57.9)	46 (56.8)
≥13 years	27 (43.5)	8 (42.1)	35 (43.2)
Gender, n (%)			
Female	30 (48.4)	10 (52.6)	40 (49.4)
Male	32 (51.6)	9 (47.4)	41 (50.6)
Race*, n (%)			
Asian	5 (8.1)	0 (0)	5 (6.2)
Black or African American	5 (8.1)	0 (0)	5 (6.2)
White	52 (83.9)	19 (100.0)	71 (87.7)
Prefer not to answer	4 (6.5)	0 (0)	4 (4.9)
Baseline convulsive seizure frequency per 28 days (N=77)†			
Median (range)	16 (4, 630.0)	21.3 (4, 2335.4)	17 (4, 2335.4)

*Multiple selections for race could be entered. †Includes n=58 and n=19 for MONARCH and ADMIRAL, respectively. One patient who received an incorrect dose in Phase 1/2a and 3 patients who had <4 seizures in Phase 1/2a baseline were excluded.

Safety and tolerability

- Overall, 59/62 patients (95.2%) in MONARCH and 19/19 patients (100%) in ADMIRAL experienced at least one treatment-emergent adverse event (TEAE) (Table 2)
- Most TEAEs were mild or moderate, and no patients withdrew from either study due to TEAEs
- TEAEs were deemed drug-related in 16 patients (25.8%) in MONARCH and in 8 patients (42.1%) in ADMIRAL, with cerebrospinal fluid protein increase and procedural vomiting being most common across both studies
- One patient from ADMIRAL who received three 70 mg doses experienced suspected unexpected serious adverse reactions but completed the study

Table 2. Summary of TEAEs*

TEAE Category Number of patients (%)	MONARCH (n=62)	ADMIRAL (n=19)	All enrolled (N=81)
Any TEAE	**59 (95.2)**	**19 (100.0)**	**78 (96.3)**
Drug-related TEAEs	16 (25.8)	8 (42.1)	24 (29.6)
Procedure-related TEAEs	35 (56.5)	8 (42.1)	43 (53.1)
≥Grade 3 TEAE	**10 (16.1)**	**3 (15.8)**	**13 (16.0)**
TESAE	**12 (19.4)**	**6 (31.6)**	**18 (22.2)**
Drug-related TESAE	0	1 (5.3)	1 (1.2)
Potential DLT	**0**	**1 (5.3)**	**1 (1.2)**
TEAE leading to study withdrawal	**0**	**0 (0)**	**0**
TEAE leading to death†	**1 (1.6)**	**0 (0)**	**1 (1.2)**

*TEAE is defined in this study as an adverse event first identified, or identified to worsen in intensity, at a time occurring after first dose of study drug.
†One patient in MONARCH died due to sudden unexpected death in epilepsy (SUDEP) that was assessed as unrelated to zorevunersen.
DLT, dose limiting toxicity; TEAE, treatment-emergent adverse event; TESAE, treatment-emergent serious adverse event.

Convulsive seizure frequency

- Patients treated with doses of 70 mg zorevunersen experienced substantial reductions in convulsive seizure frequency within the first several months after starting treatment (Figure 2)
- The majority of patients who received multiple doses of 70 mg zorevunersen experienced ≥50% reduction in convulsive seizure frequency at 3 and 6 months after their final dose, respectively (Figure 3)

Figure 2. Substantial reductions in convulsive seizure frequency from baseline*

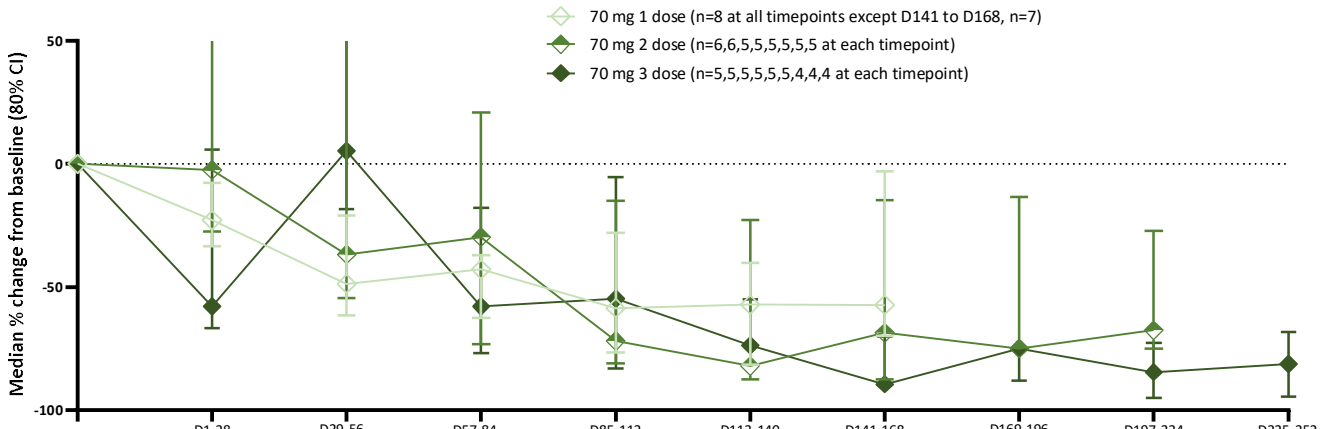
70 mg 1 dose (n=8 at all timepoints except D141 to D168, n=7)
70 mg 2 dose (n=6,6,5,5,5,5,5,5 at each timepoint)
70 mg 3 dose (n=5,5,5,5,5,5,4,4,4 at each timepoint)
Median % change from baseline (80% CI)
50
0
-50
-100
D1-28
D29-56
D57-84
D85-112
D113-140
D141-168
D169-196
D197-224
D225-252

*Zorevunersen was administered on Days 1, 29 and 57 in MONARCH, and on Days 1, 57 and 85 in ADMIRAL. MONARCH ended at Day 225 and ADMIRAL ended at Day 253. Phase 1/2a datacut was December 12, 2023 (after End of Study). One 70 mg 1-dose patient who experienced <4 seizures during the Phase 1/2a baseline period was excluded. Patients were followed for 6 months after last dose of study drug. Data were censored if <50% diary data were available for a 28-day interval (D141 to D168 for 1 patient in 70 mg 1 dose) and at time of ASM modification (1 patient in 70 mg 2 dose and 1 patient in 70 mg 3 dose)
ASM, antiseizure medication; CI, confidence interval; D, day.

Figure 3. Majority of patients treated with 70 mg zorevunersen achieved ≥50% reduction in convulsive seizure frequency

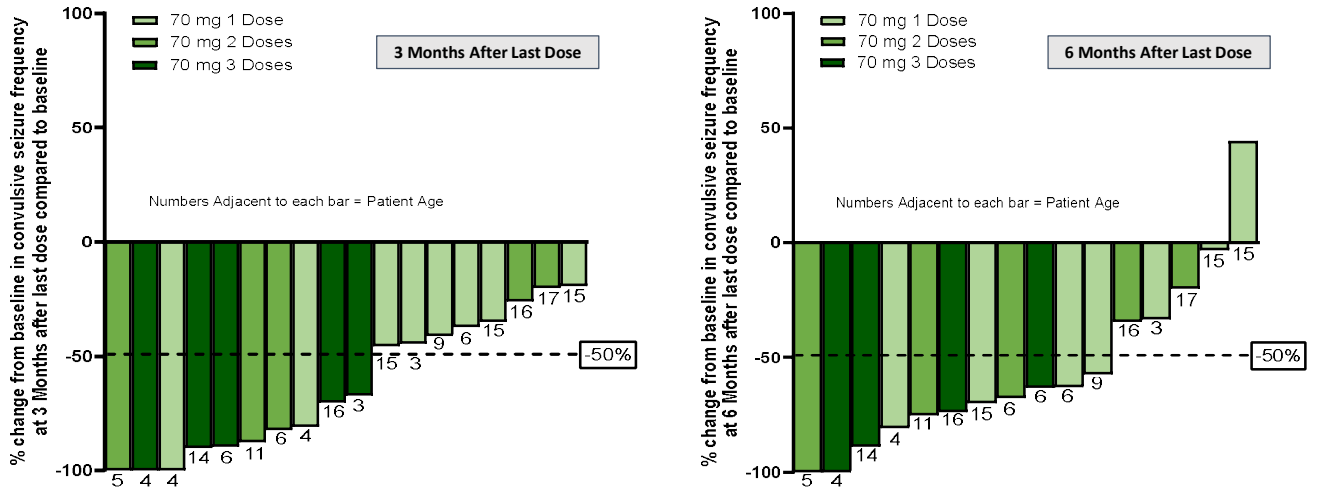
70 mg 1 Dose
70 mg 2 Doses
70 mg 3 Doses
3 Months After Last Dose
6 Months After Last Dose
Numbers Adjacent to each bar = Patient Age
% change from baseline in convulsive seizure frequency at 3 Months after last dose compared to baseline
% change from baseline in convulsive seizure frequency at 6 Months after last dose compared to baseline
-50%
100
50
0
-50
-100

Three months after last dose refers to D57 to D84 (1 dose SAD), D113 to D140 (2 dose MAD) and D141 to D168 (3 dose MAD); 6 months after last dose refers to D141 to D168 (1 dose SAD), D197 to D224 (2 dose MAD) and D225 to D252 (3 dose MAD). Phase 1/2a datacut was December 12, 2023 (after End of Study).
MAD, multiple ascending dose; SAD, single ascending dose.

Overall clinical status and quality of life

- Clinical and Caregiver Global Impression of Change (CGI-C and CaGI-C) scales indicate improvements in the overall clinical status of patients who received single or multiple doses of 70 mg zorevunersen approximately 6 months after their last dose (Figure 4)
- Patients' quality of life was improved as demonstrated by model outcomes for the EQ-VAS component of the EQ-5D-Y assessment (data not shown)

Figure 4. Improvement in overall clinical status and quality of life approximately 6 months after last dose

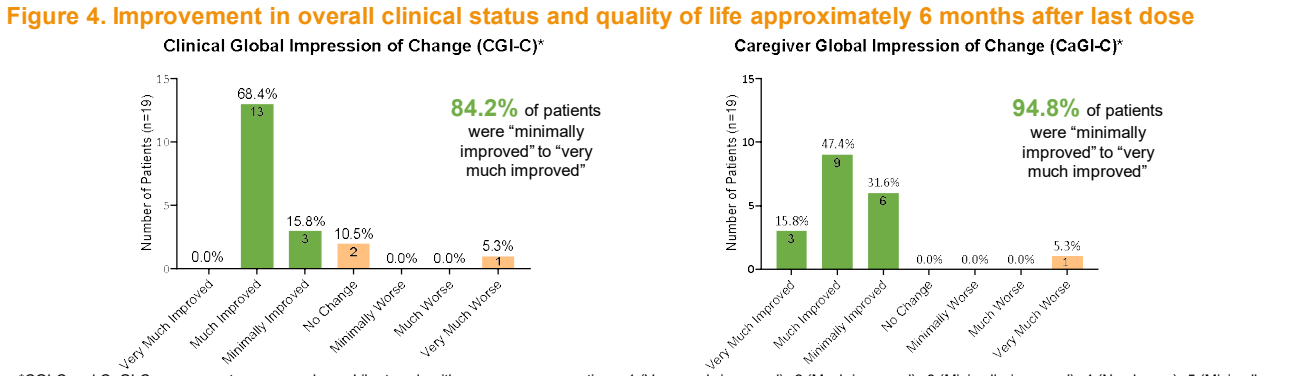
Clinical Global Impression of Change (CGI-C)*
Caregiver Global Impression of Change (CaGI-C)*
Number of Patients (n=19)
84.2% of patients were "minimally improved" to "very much improved"
94.8% of patients were "minimally improved" to "very much improved"
0.0%
68.4%
15.8%
10.5%
0.0%
0.0%
5.3%
15.8%
47.4%
31.6%
0.0%
0.0%
0.0%
5.3%
Very Much Improved
Much Improved
Minimally Improved
No Change
Minimally Worse
Much Worse
Very Much Worse

*CGI-C and CaGI-C assessments are scored on a Likert scale with seven response options: 1 (Very much improved), 2 (Much improved), 3 (Minimally improved), 4 (No change), 5 (Minimally worse), 6 (Much worse), or 7 (Very much worse). Phase 1/2a datacut was December 12, 2023 (after End of Study).
CaGI-C, Caregiver Global Impression of Change; CGI-C, Clinical Global Impression of Change.

Cognition and behavior

- The Vineland Adaptive Behavior Scales, Third Edition (Vineland-3) was included in the ADMIRAL study
- Patients in ADMIRAL experienced improvements across multiple subdomains of the Vineland-3 by end of study, approximately 9 months after starting treatment (Figure 5)

Figure 5. Improvement in adaptive behavior approximately 9 months after starting treatment with zorevunersen

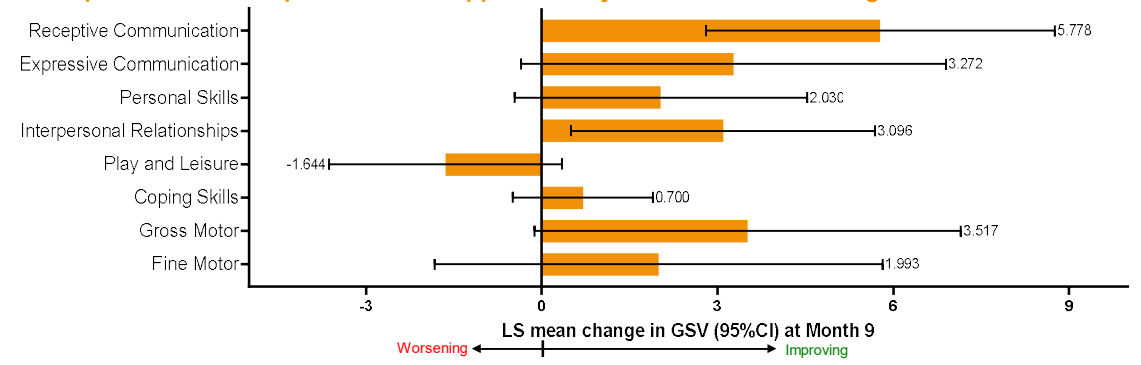
Receptive Communication
Expressive Communication
Personal Skills
Interpersonal Relationships
Play and Leisure
Coping Skills
Gross Motor
Fine Motor
5.778
3.272
2.030
3.096
-1.644
0.700
3.517
1.993
-3
0
3
6
9
LS mean change in GSV (95%CI) at Month 9
Worsening
Improving

A change of 2–3 points in GSV scores was considered meaningful by at least 50% of caregivers.[7]
Data from ADMIRAL (All Dose Cohorts) through Visit 2 (Week 16) in LONGWING from pre-treatment/naïve baseline, analyzed with Machine Learning. ADMIRAL sample size: n=18 at screen and n=17 at Week 36. ADMIRAL datacut was December 12, 2023.
CI, confidence interval; GSV, Growth Scale Value; OLE, open-label extension; Vineland-3, Vineland Adaptive Behavior Scale, Third Edition.

References: 1. Gil-Nagel A *et al. Sci Rep* 2023; 13 (1): 3355. 2. Han Z *et al. Sci Transl Med* 2020;12(558).; 3. Steel D *et al. Epilepsia* 2017; 58 (11): 1807–1816. 4. Zuberi SM *et al. Epilepsia* 2022; 63 (6): 1349–1397. 4. Villas N *et al. Epilepsy Behav* 2017; 74: 81–86; 5. Lagae L *et al. Dev Med Child Neurol* 2018; 60 (1): 62–72; 6. Sullivan J *et al.* Poster presented at the 15th European Epilepsy Congress 2024; Rome, Italy, 7–11 September 2024. 7. Condon C *et al.* Poster# 3.383 presented at the American Epilepsy Society Annual Meeting 2024; Los Angeles, USA, 6–10 December, 2024.

Acknowledgements: Studies are supported by Stoke Therapeutics, and we thank investigators, health care providers, research staff, patients, and caregivers who participated. Medical writing and editorial assistance were provided by Tanmay S. Chavan, PhD of Porterhouse Medical US and were funded by Stoke Therapeutics according to Good Publication Practice guidelines.

Presented at the American Epilepsy Society 2024 Annual Meeting • December 6–10, 2024 • Los Angeles, California, USA